P048

A Systematic Literature Review of Healthcare Burden in Patients With Behçet's Disease

Oriol Sola-Morales[1]; Sandhya Mehta[2]; Sarah Ronnebaum[3]; Erin Hart[3]; Dipen Patel[3]; Tara Nazareth[2]; Angela Padula[4]

[1]Health Institute for Technology Transfer (HITT), Barcelona, Spain; [2]Celgene Corporation, Summit, NJ, USA; [3]Pharmerit International, Bethesda, MD, USA; [4]Rheumatology Department of Lucania, San Carlo Hospital of Potenza and Madonna delle Grazie Hospital of Matera, Potenza, Italy

INTRODUCTION

- Behçet's disease (BD) is a rare systemic vasculitis disorder characterized by recurrent oral and genital ulcers, ocular disease, gastrointestinal ulcers, vascular disease, and skin lesions.[1]
- Oral ulcers are the most frequent manifestation of BD, and recurrence of oral ulcers is common throughout the disease course.[2] Organ involvement in BD may lead to irreversible damage and significant morbidity.[2,3]
- The prevalence of BD is highest in countries in the Mediterranean, Middle East, and routes extending to Eastern Asia, leading BD to be termed "Silk Road disease."[4]
- However, the prevalence of BD has increased over time across geographical areas, including developed countries (e.g., Europe), likely due to migration.[5] The change in prevalence could also be due to a change in environmental factors and increasing awareness among physicians.[2]
- Symptoms associated with BD may hinder daily activities and cause significant pain, requiring medical care, decreasing productivity, and causing emotional distress.[6] However, there is little recent evidence available to address the clinical and humanistic burden associated with BD.
- As such, we sought to understand the prevalence of BD manifestations, related symptoms, and impact on comorbidities, treatments, and quality of life (QoL).

OBJECTIVE

- To evaluate the clinical and humanistic burden among patients with BD based on a systematic literature review (SLR)

METHODS

Literature Review

- An SLR was performed to identify real-world studies reporting symptoms/manifestations, comorbidities, treatment use, and QoL among patients with BD.
 - PICOS selection criteria for inclusion of studies into the SLR are provided in **Table 1**.
- Embase/MEDLINE was searched for articles and conference abstracts published from January 1, 2003, to May 1, 2018, using a predefined search strategy combining keywords.
- Studies with patients of any age with BD from any country were included.
- Studies with <10 patients or those including only patients with BD based on severe manifestations (i.e., ocular, neurologic, or vascular involvement) or comorbidities were excluded.
- An independent researcher screened studies, identified those to be included, and extracted data. Another independent researcher performed standard quality checks to verify the results of the screening and extraction for 10% of the studies.
- For binary outcomes (i.e., symptom or manifestation prevalence, presence of comorbidities, and treatment use), the number of studies, median, interquartile range (IQR), and/or minimum-maximum (min-max) range of prevalence are reported; for continuous outcomes (i.e., QoL), the total number of comparisons and number of significantly different comparisons, as stated in the individual studies ($P \leq 0.05$), are reported.
- Weighted means were calculated for selected numerical outcomes (e.g., symptom/manifestation prevalence, percentage of patients receiving treatment) based on sample size in order to provide a summary of trends.

Table 1. PICOS Selection Criteria

	Inclusion Criteria	Exclusion Criteria
Population	• Persons (any age) with BD • Global populations	• Populations of BD patients selected based on severe manifestation or comorbidity
Intervention	• Any or none	• Not applicable
Comparators	• Any or none	• Not applicable
Outcomes	• Clinical burden – Common comorbidities – Treatment use • Humanistic burden/QoL	• Not applicable
Study design	• Observational studies (retrospective or prospective), including cohort, case-control, or cross-sectional studies • Modeling studies • Systematic reviews of cohort studies for identification of primary studies	• Trials (randomized or non-randomized) • Case series or case reports • Notes, editorials, or commentaries
Other	• Journal articles and indexed conference abstracts • Human subjects • English language • Published from January 1, 2003, to May 1, 2018 • ≥10 BD patients involved in the study	• Publication types not of interest • Animal or preclinical studies • Non-English language • Published prior to January 1, 2003

RESULTS

Summary of Included Studies

- A total of 2,656 studies were identified, of which 69 studies (56 manuscripts; 13 conference abstracts) describing patients with BD were reviewed (**Figure 1**).[7-75]

Figure 1. PRISMA Study Identification Flow Chart

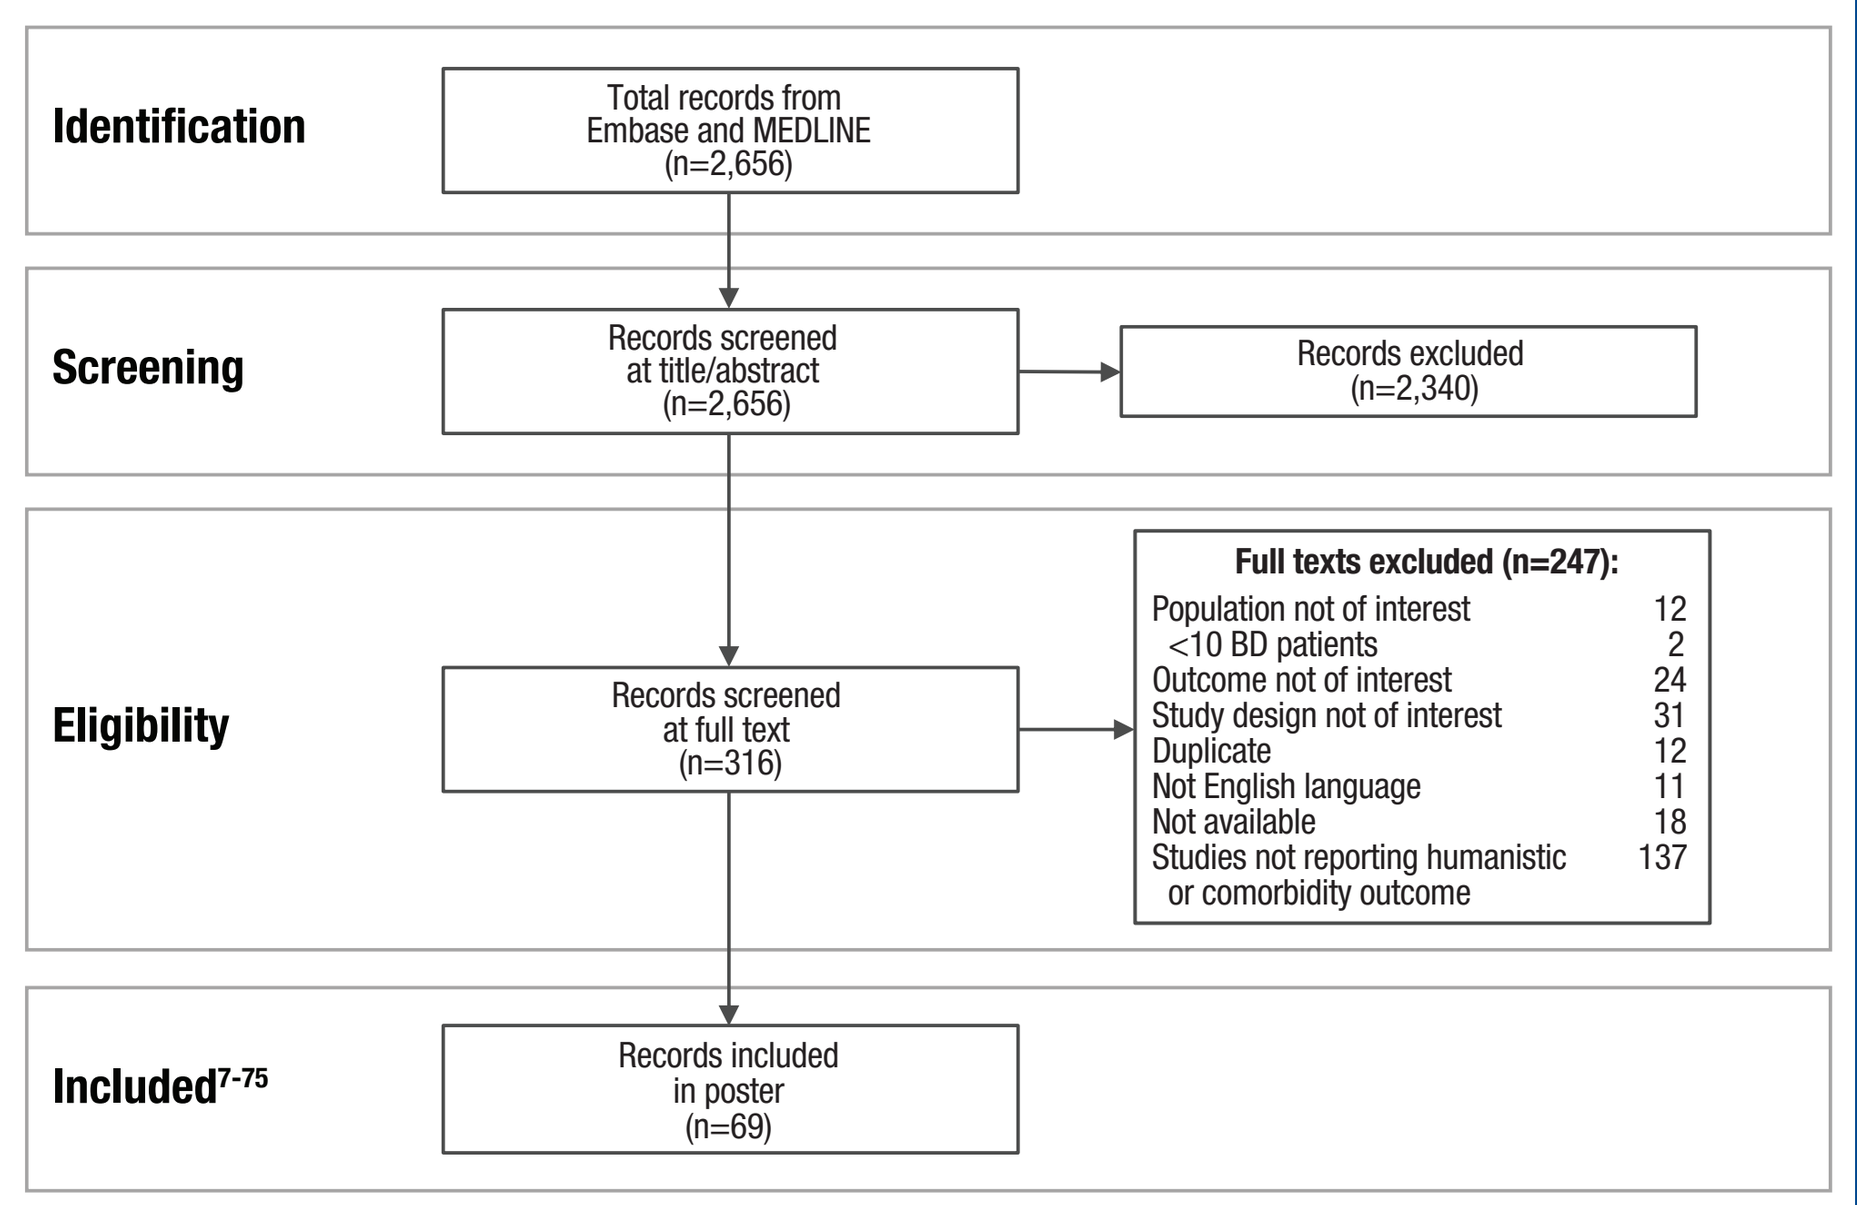

Identification
Total records from Embase and MEDLINE (n=2,656)
Screening
Records screened at title/abstract (n=2,656)
Records excluded (n=2,340)
Eligibility
Records screened at full text (n=316)
Full texts excluded (n=247):
Population not of interest 12
<10 BD patients 2
Outcome not of interest 24
Study design not of interest 31
Duplicate 12
Not English language 11
Not available 18
Studies not reporting humanistic or comorbidity outcome 137
Included[7-75]
Records included in poster (n=69)

- Within the included 69 studies, the median sample size of patients with BD was 97 (**Table 2**).
- The median age of patients at BD symptom onset was 28 years whereas it was 34 years at BD diagnosis, and approximately half of all patients with BD were male (**Table 2**).
- The majority of the data were from Turkey, reflecting 37 out of the 69 studies.

Table 2. Demographic Characteristics of Patients With BD

Characteristic	Median (IQR)	No. of Studies Reporting Outcome
BD patient sample size, n	97 (51–152)	69
Mean age, years		
At time of study	36 (35–40)	53
At BD symptom onset	28 (26–30)	18
At BD diagnosis	34 (32–39)	6
Male, %	50 (42–57)	58

Clinical Burden: Symptom Prevalence

- A total of 54 studies reported on current symptom prevalence.[7-60]
- Oral ulcers were the most prevalent manifestation in patients with BD. Among the total of 27 studies, 19 studies reported an oral ulcer prevalence of 100% (IQR: 84%–100%) (**Table 3**).
- One study reported an average of 12.7 oral ulcers per patient per year.[48] In another study, oral ulcers were the only manifestation currently or previously experienced by all patients.[60]
- More than 50% of all BD patients reported experiencing other symptoms. The top 5 prevalent symptoms included fatigue or sleep-related issues, pain/neuropathic pain syndrome/myalgia, skin lesions, headache/migraine, and arthritis, indicating that oral ulcers could often be experienced concurrently with other BD symptoms (**Table 3**).

Table 3. Symptom or Manifestation Prevalence in Patients With BD

Symptom or Manifestation	No. of Studies Reporting Symptom	Prevalence, %, Median (IQR)	Prevalence, %, Weighted Mean
Oral ulcer*	27	100 (84–100)	84
Fatigue or sleep-related issues	7	69 (52–78)	74
Pain/neuropathic pain syndrome/myalgia	4	59 (39–63)	49
Any skin lesion*	19	58 (48–81)	58
Headache/migraine*	5	58 (55–61)	54
Arthritis*,†	27	48 (38–72)	53
Other ocular manifestations*	24	36 (32–44)	38
Erythema nodosum*	9	33 (20–42)	31
Any eye lesion*	9	29 (24–41)	29
Any gastrointestinal lesion	20	24 (8–39)	17
Vascular/vessel involvement*	17	16 (11–22)	17
Blood clots‡	11	11 (8–12)	7
Any neurologic symptom*	25	9 (4–20)	17
Gastrointestinal manifestations*	2	7 (3–19)	25

Note: The red box indicates symptoms or manifestations affecting >50% of the population (weighted mean).
*Included in the Behçet's Disease Current Activity Form, which is a BD-specific clinical instrument that scores the history of 12 clinical features experienced within the 4 weeks prior to the day of assessment.[76] †Arthritis includes arthritis; arthralgia; joint problems; articular, rheumatologic, or musculoskeletal involvement; and other arthritic manifestations.
‡Blood clots include cerebral venous thrombosis, vena cava thrombosis, thrombosis, and thrombophlebitis.

Clinical Burden: Comorbidities

- Twenty-six studies reported on comorbidities present among patients with BD.[7,8,10-12,15-17,24,29,35,36,38,40,43,45,50,59,62,64-70]
- Several comorbidities were significantly higher in patients with BD vs healthy controls, including emotional issues (4 studies; 9%–33% difference), fibromyalgia (4 studies; 3%–17% difference), and hearing loss (3 studies; 10%–39% difference) (**Figure 2**).[7,12,16,17,29,36,40,43,59,67,69]

Figure 2. Comorbidity Comparison: Percent Difference in Prevalence Between Patients With BD and Healthy Controls*

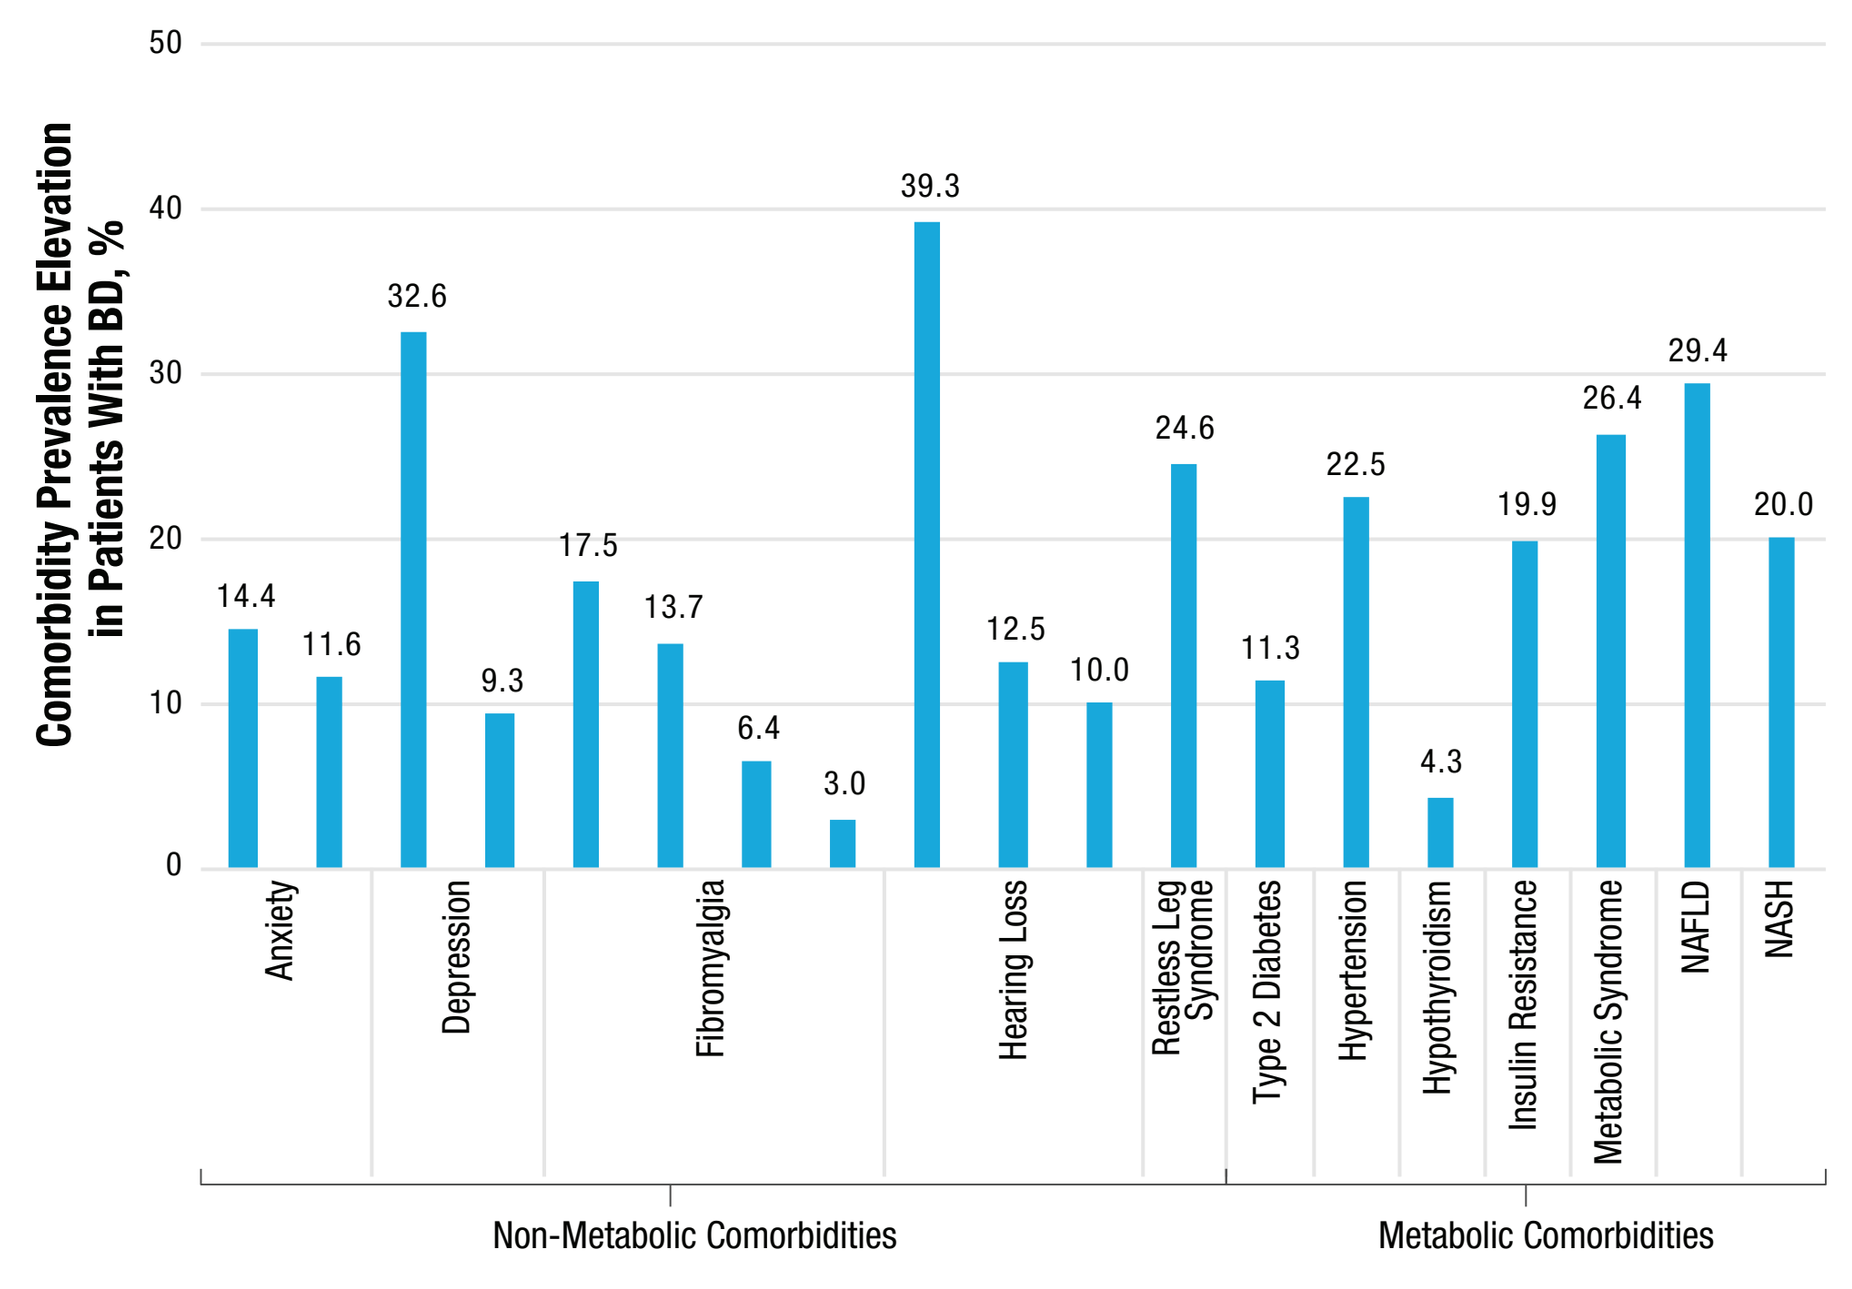

Comorbidity Prevalence Elevation in Patients With BD, %
0
10
20
30
40
50
14.4
11.6
32.6
9.3
17.5
13.7
6.4
3.0
39.3
12.5
10.0
24.6
11.3
22.5
4.3
19.9
26.4
29.4
20.0
Anxiety
Depression
Fibromyalgia
Hearing Loss
Restless Leg Syndrome
Type 2 Diabetes
Hypertension
Hypothyroidism
Insulin Resistance
Metabolic Syndrome
NAFLD
NASH
Non-Metabolic Comorbidities
Metabolic Comorbidities

*Each bar represents 1 study. Percent difference is calculated by subtracting the percentage of healthy controls with a comorbidity from the percentage of patients with BD and a comorbidity.
NAFLD=non-alcoholic fatty liver disease; NASH=nonalcoholic steatohepatitis.

Clinical Burden: Treatment Patterns

- Twenty-four studies reported on treatment patterns among patients with BD.[25,26,30,31,34,37,38,41,43-46,48,50-52,54,55,58-63]
- Patients with BD are treated with a variety of predominantly off-label treatments, most commonly colchicine, which is recommended first line for the prevention of recurrent mucocutaneous lesions (**Table 4**).[3]
- A weighted average of 11% of patients with BD did not receive treatment at the time of study; reasons included recent diagnosis, lack of compliance, or lack of need for drug.

Table 4. Treatment Patterns Among Patients With BD

Treatment	No. of Studies Reporting Treatment Use	Prevalence, %, Median (IQR)	Prevalence, %, Weighted Mean
Colchicine	19	52 (40–87)	45
DMARD*	2	49 (36–56)	38
Steroid (any)	17	41 (18–53)	34
Other†	8	22 (7–38)	29
Azathioprine	7	15 (13–31)	19
Other immunosuppressants	16	9 (4–31)	15
None	10	9 (8–15)	11
Anti-TNF	7	5 (3–9)	6
Interferon	2	5 (3–6)	4
Methotrexate	3	3 (2–4)	3

*"DMARD" provided by study; no further information was given on the particular therapy. †"Other" includes anticoagulation therapy, "cytotoxic drugs," hydroxyquinolone, metformin, non-steroidal anti-inflammatory drugs, pentoxyphilline, sulphasalazine, or reported as "other" in the study.
DMARD=disease-modifying antirheumatic drug; TNF=tumor necrosis factor.

Humanistic Burden

- Forty-two studies reported humanistic outcomes among patients with BD.[13-22,24-26,28-31,33,34,37,38,41,44,46,51,53,55,57,58,60,62-64,67-69,71-75]
- Patients with BD were compared by study authors against healthy controls (22 studies) and against other chronic inflammatory conditions, such as rheumatoid arthritis, systemic lupus erythematosus, ankylosing spondylitis, recurrent aphthous stomatitis, and psoriasis (7 studies).
- Compared with healthy controls, patients with BD had significantly worse QoL overall and across many domains, including physical/social functioning, sleep/fatigue, and mental health, in 80% (74 of the 92) of comparisons made (**Figure 3**).
- Compared with other chronic inflammatory conditions, patients with BD had similar or significantly worse QoL in 84% (16 of the 19) of comparisons made (**Figure 3**).

Figure 3. Summary of Humanistic Burden Impairments in Patients With BD vs Healthy Controls and in Patients With Inflammatory Conditions

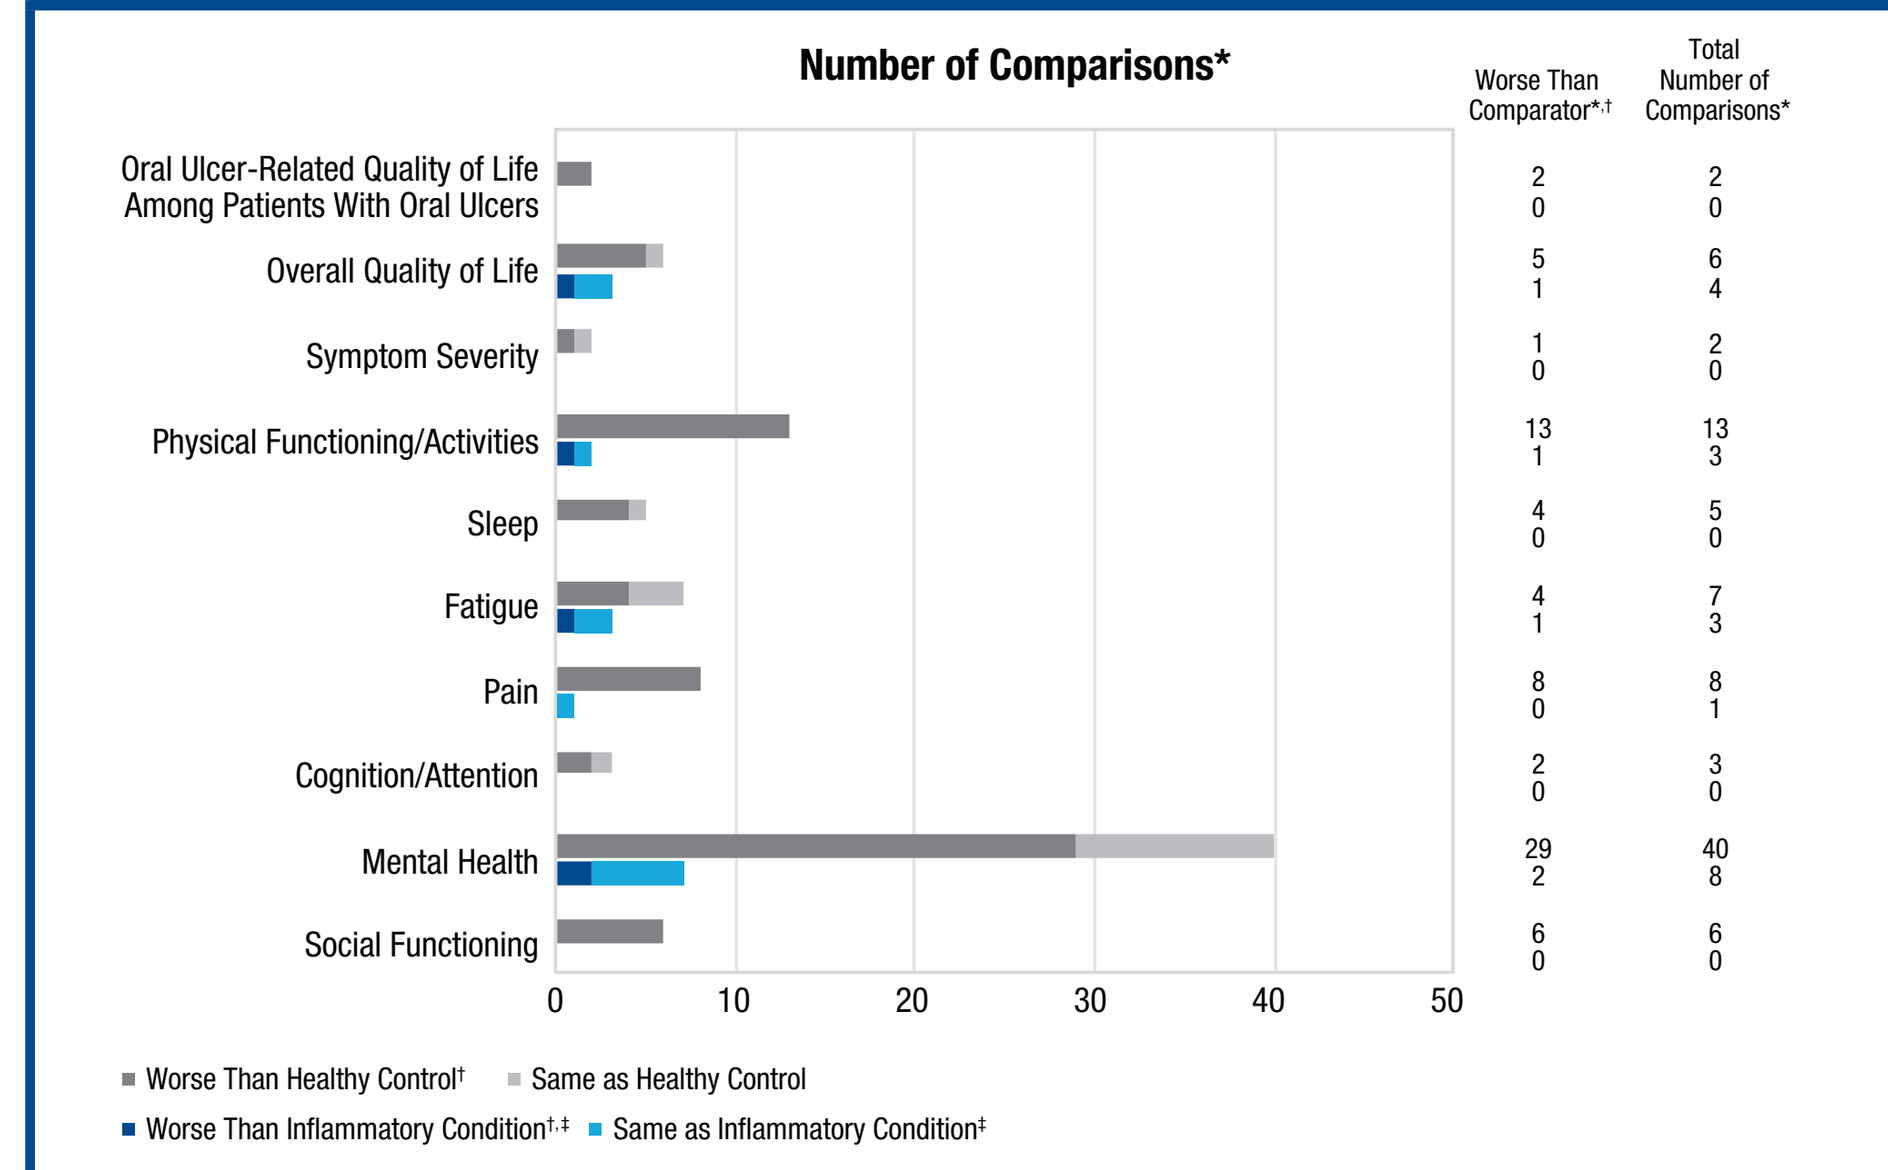

Number of Comparisons*
Worse Than Comparator*,†
Total Number of Comparisons*
Oral Ulcer-Related Quality of Life Among Patients With Oral Ulcers 2 0 2 0
Overall Quality of Life 5 1 6 4
Symptom Severity 1 0 2 0
Physical Functioning/Activities 13 1 13 3
Sleep 4 0 5 0
Fatigue 4 1 7 3
Pain 8 0 8 1
Cognition/Attention 2 0 3 0
Mental Health 29 2 40 8
Social Functioning 6 0 6 0
0
10
20
30
40
50
Worse Than Healthy Control†
Same as Healthy Control
Worse Than Inflammatory Condition†,‡
Same as Inflammatory Condition‡

*Because most studies report QoL using >1 instrument, the total number of comparisons is reported instead of the total number of studies. †"Worse" is defined as significantly different ($P<0.05$). ‡Inflammatory conditions include rheumatoid arthritis, systemic lupus erythematosus, ankylosing spondylitis, recurrent aphthous stomatitis, and psoriasis.

- Compared with healthy controls, dental patients, and benign oral ulcer patients, patients with BD who have oral ulcers showed significant impairment in oral health-related QoL and overall QoL (**Table 5**).[47,63]

Table 5. Oral Ulcer-Related QoL Studies and Scores

Study	Country	Population	Oral Ulcer Prevalence, %	Value
Oral Health-Related QoL (16–144, with lower scores indicating worse QoL)				
Mumcu, 2006[63]	Turkey	All BD*	100	42.0†
		Healthy controls	NR	48.7†
		Dental patients	NR	43.6†
		Recurrent aphthous stomatitis	NR	50.1†
Oral Health Impact Profile (0–56, with higher scores indicating worse QoL)				
Mumcu, 2006[63]	Turkey	All BD*	100	20.5‡
		Healthy controls	NR	12.3‡
		Dental patients	NR	15.1‡
		Recurrent aphthous stomatitis	NR	15.3‡
Mumcu, 2009[47]	Turkey and United Kingdom	All BD – United Kingdom All BD – Turkey	100 100	22.7 20.4

*BD patients scored significantly lower ($P<0.05$) than healthy controls in all 8 domains of the 36-Item Short-Form Health Survey. †$P<0.05$ across groups by analysis of variance. ‡$P<0.05$ across groups by the Kruskal-Wallis test.
NR=not reported.

STRENGTHS AND LIMITATIONS

Strengths

- This is the first SLR to comprehensively describe the clinical and humanistic burden of patients with BD.

Limitations

- There was disproportionate reporting of data from Turkey; data from other countries where BD is relatively prevalent (e.g., Jordan, Lebanon, Israel, Saudi Arabia, China, Japan, France) were limited to 0 or 1 study per country.
- Most studies (77%) identified for inclusion were cross-sectional, which does not facilitate a longitudinal view of the relapsing and progressive nature of BD symptoms and its impact on humanistic burden.
- Patient characteristics were not always clarified.
- Heterogeneity was evident across the QoL assessment instruments used.
- Medication use and QoL outcomes by manifestation were not clearly described.

CONCLUSIONS

- Patients with BD experience an increased symptom, comorbidity, and humanistic burden of illness compared with healthy controls.
- Patients with BD experience comparable or worse humanistic burden of illness compared with patients with other chronic inflammatory conditions, such as rheumatoid arthritis, systemic lupus erythematosus, ankylosing spondylitis, recurrent aphthous stomatitis, and psoriasis.
- Findings confirm that oral ulcers are highly prevalent, often manifest concurrently with a number of other symptoms, and are directly associated with poor QoL. Patients with oral ulcers associated with BD therefore represent a significant proportion of BD patients overall and have important unmet needs.
- Currently, treatment options are lacking for patients with oral ulcers associated with BD and are predominantly off-label. A recent SLR highlighted the lack of robust clinical trials and the varied results associated with current treatments.[77]
- New treatments for patients with oral ulcers associated with BD should demonstrate robust clinical efficacy (i.e., sustained control of oral ulcer occurrence), pain reduction, and QoL improvements.

REFERENCES

1. Tong B, et al. *Front Immunol.* 2019;10:665. **2.** Davatchi F, et al. *Exp Rev Clin Immunol.* 2017;13:57–65. **3.** Hatemi G, et al. *Ann Rheum Dis.* 2018;77:808-818. **4.** Maldini C, et al. *Rheumatology (Oxford).* 2018;57:185-195. **5.** Mahr A, et al. *Arthritis Rheum.* 2008;58:3951-3959. **6.** McNally TW, et al. *Orphanet J Rare Dis.* 2017;12:130. **7.** Abdelraheem T, et al. *Acta Reumatol Port.* 2013;38:252-260. **8.** Abou-Raya A, et al. *Ann Rheum Dis.* 2014;73(Suppl 2). Abstract FRI0448. **9.** Ahn JK, et al. *Rheumatol Int.* 2010;30:831-835. **10.** Akman A, et al. *J Clin Periodontol.* 2007;34:485-491. **11.** Alaoui FZ, Moudatir M. *Ann Rheum Dis.* 2017;76:322. **12.** Al-Izzi MK, Jabber AS. *J Med Liban.* 2004;52:86-90. **13.** Bernabé E, et al. *Rheumatology.* 2010;49:2165-2171. **14.** Bodur H, et al. *Clin Rheumatol.* 2006;25:329-333. **15.** Borhani Haghighi A, et al. *Iran J Med Sci.* 2007;32:110-113. **16.** Buyuktas D, et al. *Clin Exp Rheumatol.* 2015;33:107-112. **17.** Can M, et al. *Arthritis Rheum.* 2012;64:S1008-S1009. **18.** Canpolat Ö, Yurtsever S. *Asian Nurs Res (Korean Soc Nurs Sci).* 2011;5:229-235. **19.** Dursun R, et al. *Int J Psychiatry Clin Pract.* 2007;11:16-20. **20.** Ertam I, et al. *Clin Exp Rheumatol.* 2009;27:S18-S22. **21.** Evcik D, et al. *Clin Rheumatol.* 2013;32:33-36. **22.** Fabiani C, et al. *Intern Emerg Med.* 2017;12:947-955. **23.** Fatemi A, et al. *Int J Rheum Dis.* 2017;20:97-102. **24.** Garip Y, et al. *Turk Fiz Tip Rehab D.* 2016;62:45-50. **25.** Güler T, et al. *Turkiye Klinikleri J Med Sci.* 2017;37:130-135. **26.** Gur A, et al. *Clin Rheumatol.* 2006;25:524-531. **27.** Haghighi AB, et al. *Headache.* 2008;48:424-429. **28.** Hatemi G, et al. *Rheumatology.* 2011;50:ii26-ii27. **29.** Ilhan B, et al. *Int J Rheum Dis.* 2018;21:2139-2145. **30.** Ishchenko D, et al. *Ann Rheum Dis.* 2015;74:1121-1122. **31.** Karlidag R, et al. *J Eur Acad Dermatol Venereol.* 2003;17:670-675. **32.** Kilic H, et al. *Clin Exp Rheumatol.* 2010;28:S136-S137. **33.** Kister I, et al. *Clin Exp Rheumatol.* 2010;28:S112-S113. **34.** Koca I, et al. *Clin Rheumatol.* 2015;34:1259-1263. **35.** Krause L, et al. *Graefes Arch Clin Exp Ophthalmol.* 2009;247:661-666. **36.** Kulahli I, et al. *Hear Res.* 2005;203:28-31. **37.** Kural-Seyahi E, et al. *Medicine.* 2003;82:60-76. **38.** Lee J, et al. *Korean J Intern Med.* 2017;32:352-359. **39.** Lin J, et al. *Yan Ke Xue Bao.* 2004;20:155-157. **40.** Mahdi B, et al. *Otolaryngol Head Neck Surg.* 2007;137:439-442. **41.** Melikoğlu M, Melikoglu MA. *Acta Reumatol Port.* 2014;39:46-53. **42.** Melikoglu MA, Melikoglu M. *Rheumatol Int.* 2010;30:941-946. **43.** Meric A, et al. *B-ENT.* 2015;11:31-37. **44.** Monastero R, et al. *J Neurol Sci.* 2004;220:99-104. **45.** Mumcu G, et al. *Rheumatology (Oxford).* 2004;43:1028-1033. **46.** Mumcu G, et al. *Turk J Med Sci.* 2017;47:535-541. **47.** Mumcu G, et al. *J Oral Pathol Med.* 2009;38:406-409. **48.** Naito M, et al. *Genet Res Int.* 2014;2014:9303048. **49.** Ozguler Y, et al. *Ann Rheum Dis.* 2013;72(Suppl 3). Abstract AB0454. **50.** Saadoun D, et al. *Arthritis Care Res.* 2009;61:518-526. **51.** Saygin C, et al. *Clin Exp Rheumatol.* 2015;33:30-35. **52.** Senusi A, et al. *Orphanet J Rare Dis.* 2015;10:117. **53.** Senusi AA, et al. *Clin Exp Rheumatol.* 2017;35(Suppl 108):43-50. **54.** Sibley C, et al. *J Rheumatol.* 2014;41:1379-1384. **55.** Sücüllü Karadağ Y, et al. *J Neurol Sci.* 2014;31:511-520. **56.** Sut N, et al. *Rheumatology.* 2007;46:678-682. **57.** Tascilar NF, et al. *J Sleep Res.* 2012;21:281-288. **58.** Uğuz F, et al. *Gen Hosp Psychiatry.* 2007;29:21-24. **59.** Ulaşoğlu C, et al. *Nobel Med.* 2013;9:35-42. **60.** Yazmalar L, et al. *Int J Rheum Dis.* 2017;20:2062-2069. **61.** Han M, et al. *J Gastroenterol.* 2017;52:920-928. **62.** Mehta P, et al. *Ann Rheum Dis.* 2013;72(Suppl 3). Abstract SAT0170. **63.** Mumcu G, et al. *Oral Dis.* 2006;12:145-151. **64.** Pandey A, et al. *Cardiology (Switzerland).* 2014;129:203-206. **65.** Lee SS, et al. *Clin Exp Rheumatol.* 2005;23:S15-S19. **66.** Melikoglu M, Melikoglu MA. *Rheumatol Int.* 2013;33:1219-1222. **67.** Toprak M, et al. *Turk Fiz Tip Rehab D.* 2017;63:160-164. **68.** Ovcharov P, et al. *Ann Rheum Dis.* 2017;76:616-617. **69.** Ediz L, et al. *J Int Med Res.* 2011;39:759-765. **70.** Ene P, et al. *Ann Rheum Dis.* 2016;75:1216. **71.** Alder NM, et al. *Clin Exp Rheumatol.* 2008;26:S110-S113. **72.** Can M, et al. *Ann Rheum Dis.* 2013;71(Suppl 3). Abstract FRI0213. **73.** Dutra LA, et al. *J Neurol Sci.* 2013;327:1-5. **74.** Singh J, et al. *Clin Exp Rheumatol.* 2010;28:S159. **75.** Taner E, et al. *Int J Dermatol.* 2007;46:1118-1124. **76.** International Society for Behcet's Disease; 2006. Presented at: http://www.behcetdiseasesociety.org/behcetwsData/Uploads/files/BehcetsDiseaseActivityForm.pdf. **77.** Leccese P, et al. *Semin Arthritis Rheum.* 2019;48:752-762.

ACKNOWLEDGEMENTS

The authors acknowledge financial support for this study from Celgene Corporation. The authors received editorial support in the preparation of this poster from Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, sponsored by Celgene Corporation, Summit, NJ, USA. The authors, however, directed and are fully responsible for all content and editorial decisions for this poster.

CORRESPONDENCE

Oriol Sola-Morales – osola@hittbcn.com

DISCLOSURES

OS-M: EUCOPE – grant/research support; Bayer, Novartis, Pfizer, and Sanofi – speakers bureau; Celgene Corporation – consultant, honoraria, and advisor. **SM:** Celgene Corporation – employment. **SR, EH, & DP:** Pharmerit International – employment. **TN:** Celgene Corporation – employment. **AP:** Eli Lilly, EMS Italia – speakers bureau.

Copies of this poster obtained through the QR Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at: the European Congress of Rheumatology EULAR 2019; 12–15 June 2019; Madrid, Spain.